PIN75

A NEW COST-EFFECTIVENESS MODELLING APPROACH IN CHRONIC HEART FAILURE WITH REDUCED EJECTION FRACTION

Trueman D[1], McMurray JJV[2], Cowie MR[3], Cohen A[4], Briggs A[2], Taylor M[5], de Pouvourville G[6], Hancock E[1], Mumby-Croft J[1], Haroun R[7], Deschaseaux C[7]

[1]DRG Abacus, Bicester, UK; [2]University of Glasgow, Glasgow, UK; [3]Imperial College London, London, UK; [4]Hôpitaux Universitaires Paris-Est, Paris, France; [5]York Health Economics Consortium Ltd., York, UK; [6]ESSEC Business School, Cergy-Pontoise, France; [7]Novartis Pharma AG, Basel, Switzerland.

BACKGROUND

- Deterioration of heart function in patients with heart failure and reduced ejection fraction (HFrEF) is chronic and progressive; as such, treatment aims to prevent or slow the worsening of heart function in order to reduce mortality, hospitalisation and symptoms.[1,2]
- The current first-line treatment for the management of HFrEF is an angiotensin-converting enzyme inhibitor (ACEi) in combination with a beta-blocker (BB), augmented with mineralocorticoid receptor antagonists (MRA) where symptoms persist.[1-3]
- Sacubitril/valsartan (LCZ696) is a novel oral therapy for the treatment of patients with HFrEF and has demonstrated superior efficacy vs the ACEi enalapril in a large, phase III, double-blind, randomised controlled trial (PARADIGM-HF).[4]

OBJECTIVE

- The objective of this study was to develop a model framework for evaluating the cost-effectiveness of sacubitril/valsartan compared with ACEi in the treatment of adult patients with HFrEF from the perspective of the healthcare provider.

METHODS

- A systematic literature review was performed [5]. Searches were conducted in MEDLINE, EMBASE, EconLit, and Cochrane Library databases, with supplementary hand searching of conferences and HTA websites.
- Of 63 distinct analyses identified, 33 used decision-analytic models. Structures were most commonly described as Markov models (n=27), but health state definitions varied.
- The health states most frequently described were 'alive' and 'dead', with outcomes such as hospitalisation or New York Heart Association (NYHA) class distribution commonly considered within the 'alive' state to predict cost and quality of life effects.
- A de novo model structure was then developed, which aimed to use the most common and appropriate features observed in published models. It was also designed to reflect the primary trial analysis, to best capture natural history and to facilitate the investigation of patient populations with different characteristics at baseline.

RESULTS

- The model is structured as a two-state Markov model with health states 'alive' and 'dead', and hospitalisation, adverse events and health-related quality of life (HRQoL) modelled within the 'alive' state.
- Regression models are used to predict mortality, hospitalisation and EQ-5D utility.
 - The regression models control for baseline characteristics of patients; this approach facilitates estimation of different absolute event rates and HRQoL based on alternative patient characteristics and allows for extrapolation beyond the trial.
- The model is run once using the baseline characteristics of each patient in PARADIGM-HF, and the resulting outcomes averaged across the entire cohort. This allows for the characterisation of cost-effectiveness across a heterogeneous cohort in the presence of non-linearities.
- This approach also facilitates analyses in which the PARADIGM-HF population is re-weighted to better reflect local HF populations.

Table 1: Model summary

Evaluation component	Description
Population	Adults with symptomatic heart failure and reduced ejection fraction; subgroups considered
Analysis	Cost-utility
Pespective	Healthcare provider
Time horizon	Lifetime
Structure	Two-state Markov model ('alive' and 'dead') with hospitalisation, adverse events and HRQoL modelled within the 'alive' state
Cycle length	1 month (with half-cycle correction)
Included events	Mortality, hospitalisation, adverse events
Included costs	Primary and background pharmacological therapies, hospitalisation, monthly resource use and adverse events
Key data sources	Clinical and HRQoL data, and baseline characteristics taken from PARADIGM-HF, monthly resource use from an analysis of CPRD and costs from published national sources

Abbreviations: HRQoL, health-related quality of life | CPRD, Clinical Practice Research Datalink.

- Regression models were derived from PARADIGM-HF, a double-blind phase III trial comparing sacubitril/valsartan (200mg bid) with the ACEi enalapril (10mg bid) in 8399 patients with HFrEF.
- Mortality is predicted using a multivariable parametric survival model assuming a Gompertz distribution, which provided the most conservative estimate of survival.
 - Previous economic evaluations in HF have observed mortality to be the main determinant of cost-effectiveness.[6]
 - Two scenarios for predicting mortality rates were therefore considered: 1) all-cause mortality estimated via a parametric survival model (with treatment arm and baseline characteristics as independent variables) and 2) CV mortality estimated via a parametric survival model with non-CV mortality informed by UK life-tables adjusted to remove CV mortality.
- Hospitalisation rates are estimated using a negative binomial regression model with treatment arm and baseline characteristics as independent variables.
- HRQoL is estimated using a mixed model to predict EQ-5D score as a function of baseline characteristics, treatment arm, hospitalisation, adverse events (AEs) and time from randomisation.
 - This represents a novel approach in which change in EQ-5D over time is explicitly modelled. Previous studies have predicted EQ-5D as a function of NYHA and assumed that NYHA remains constant from the end of trial follow-up to death.
- Although hospitalisation is expected to incorporate the costs of serious AEs, the costs of less serious AEs are also considered assuming a constant rate for each. A simplistic approach was considered reasonable, as the low associated costs are not a driver of cost-effectiveness. AEs included were all those pre-specified in PARADIGM-HF and reported by McMurray et al [13].

CONCLUSIONS

- The new framework builds on decision-analytic models previously developed in heart failure, with two key developments:
 - The approach in which individual subjects are exposed to multivariable risk equations allows for the accurate estimation of model outcomes in a heterogeneous cohort, and the exploration of cost-effectiveness across subgroups and alternative patient populations.
 - The novel approach in which HRQoL is estimated directly as a function of time provides a more parsimonious model with improved clinical plausibility compared with other structures in the literature.

Figure 1: Conceptual model

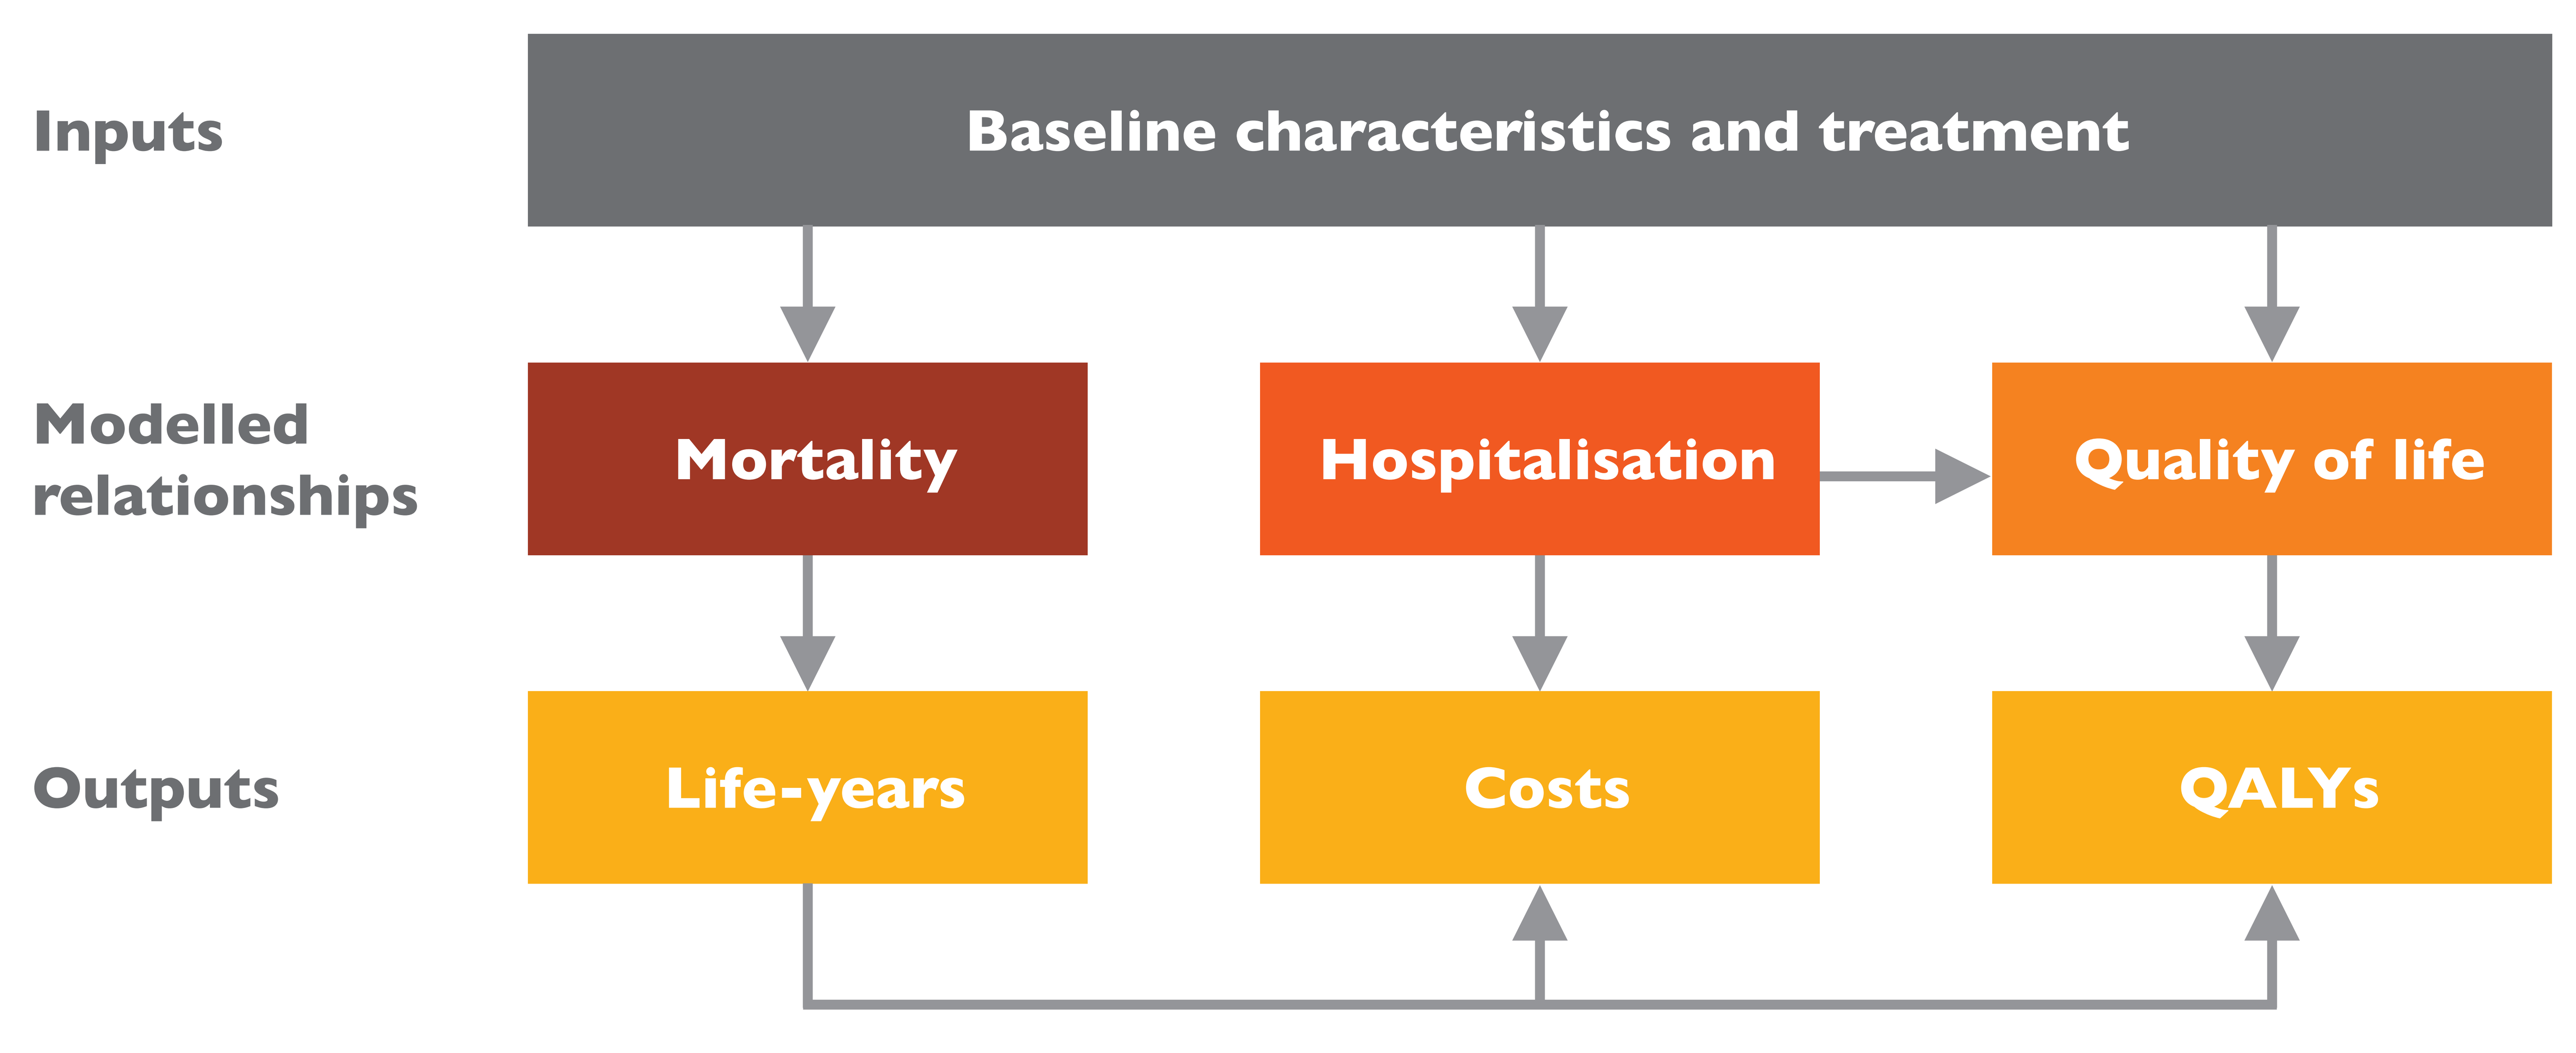
Inputs
Baseline characteristics and treatment
Modelled relationships
Mortality
Hospitalisation
Quality of life
Outputs
Life-years
Costs
QALYs

REFERENCES

1. European Society of Cardiology. ESC Guidelines for the diagnosis and treatment of acute and chronic heart failure 2012. Available at: http://eurheartj.oxfordjournals.org/content/33/14/1787.full.pdf (last accessed: 28 August 2014). 2012.
2. National Institute for Health and Care Excellence. Chronic Heart Failure (Clinical Guideline 108). Available at: http://www.nice.org.uk/guidance/CG108 (last accessed: 28 August 2014). 2010.
3. Scottish Intercollegiate Guidelines Network. SIGN Guideline 95: Management of chronic heart failure. Available at: http://www.sign.ac.uk/guidelines/fulltext/95/section1.html (last accessed: 28 August 2014).
4. McMurray JJ, Packer M, Desai AS, et al. Angiotensin-neprilysin inhibition versus enalapril in heart failure. The New England journal of medicine. Sep 11 2014;371(11):993-1004.
5. Webb N, Cowie MR, Taylor M, et al. The cost-effectiveness of treatment for chronic heart failure: a systematic review. ISPOR 18th Annual European Congress. PCV101. 2015.
6. Griffiths A, Paracha N, Davies A, Branscombe N, Cowie MR, Sculpher M. The cost effectiveness of ivabradine in the treatment of chronic heart failure from the UK National Health Service perspective. Heart. Mar 14 2014.